In Vitro Characterization of Gene Silencing Methods With the Potential to Treat Autosomal Dominant Hearing Loss

Katherine D. Gribble[1], Robert Ng[1], Priya Bansode[1], Danielle R. Lenz[1], Gregory S. Robinson[1]

[1]AKOUOS, INC., BOSTON, MA

INTRODUCTION

- Autosomal dominant hearing loss (ADHL) is estimated to represent 30% of all genetic forms of hearing loss and is caused by mutation in one copy of the gene; most individuals affected by ADHL harbor toxic gain-of-function and/or dominant negative mutations
- Current interventions, including hearing aids and cochlear implants, do not address the underlying genetic cause of hearing loss
- The development of adeno-associated viral (AAV) vectors as an intracochlear drug delivery platform poses a unique opportunity in the potential treatment of genetic forms of hearing loss by developing precision medicines that are selectively designed to target the affected gene
- Here we present efforts to screen potential candidates for the potential treatment of ADHL using AAV vector delivering RNAi biallelic gene silencing or CRISPR/Cas9 biallelic gene editing techniques, with simultaneous add-back of the functional copy of the gene of interest

FIGURE 1: Overview of genetically-driven hearing loss

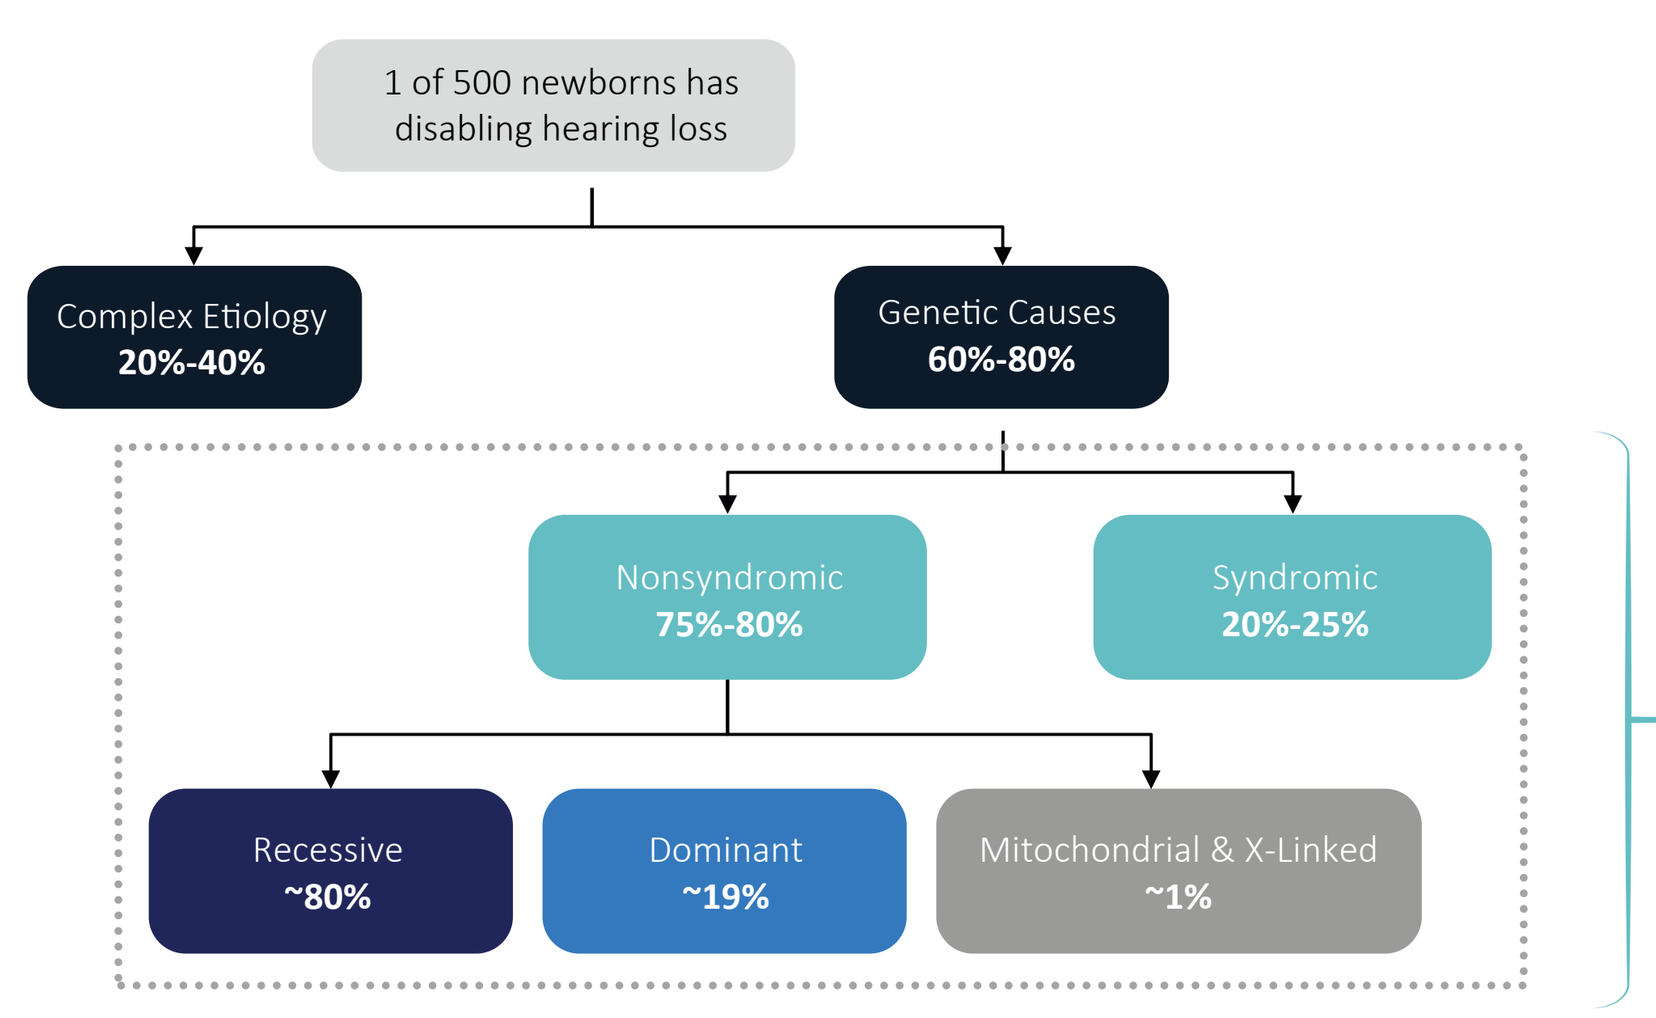
1 of 500 newborns has disabling hearing loss
Complex Etiology 20%-40%
Genetic Causes 60%-80%
Nonsyndromic 75%-80%
Syndromic 20%-25%
Recessive ~80%
Dominant ~19%
Mitochondrial & X-Linked ~1%

Note: Syndromic hearing loss means, in addition to hearing loss, the individual has other symptoms of medical importance.

	Nonsyndromic	Syndromic
Autosomal Recessive	>75 genes	>30 genes
Autosomal Dominant	>48	>10

References: Sloan-Heggen et al 2016 Hum Genet (2016); Morton et al N Engl J Med.(2006); Sheffield et al Cold Spring Harb Perspect Med.(2019); Kral et al N Engl J Med. (2010)

METHODS

- In vitro screening of engineered microRNA plasmids using Gene of Interest mScarlet reporter plasmid and Dual Luciferase reporter plasmids
- Evaluate Gene of Interest protein knockdown using fluorescence imaging and Western Blot
- Evaluate guide/passenger strand generation and knockdown using Dual Luciferase reporter assay

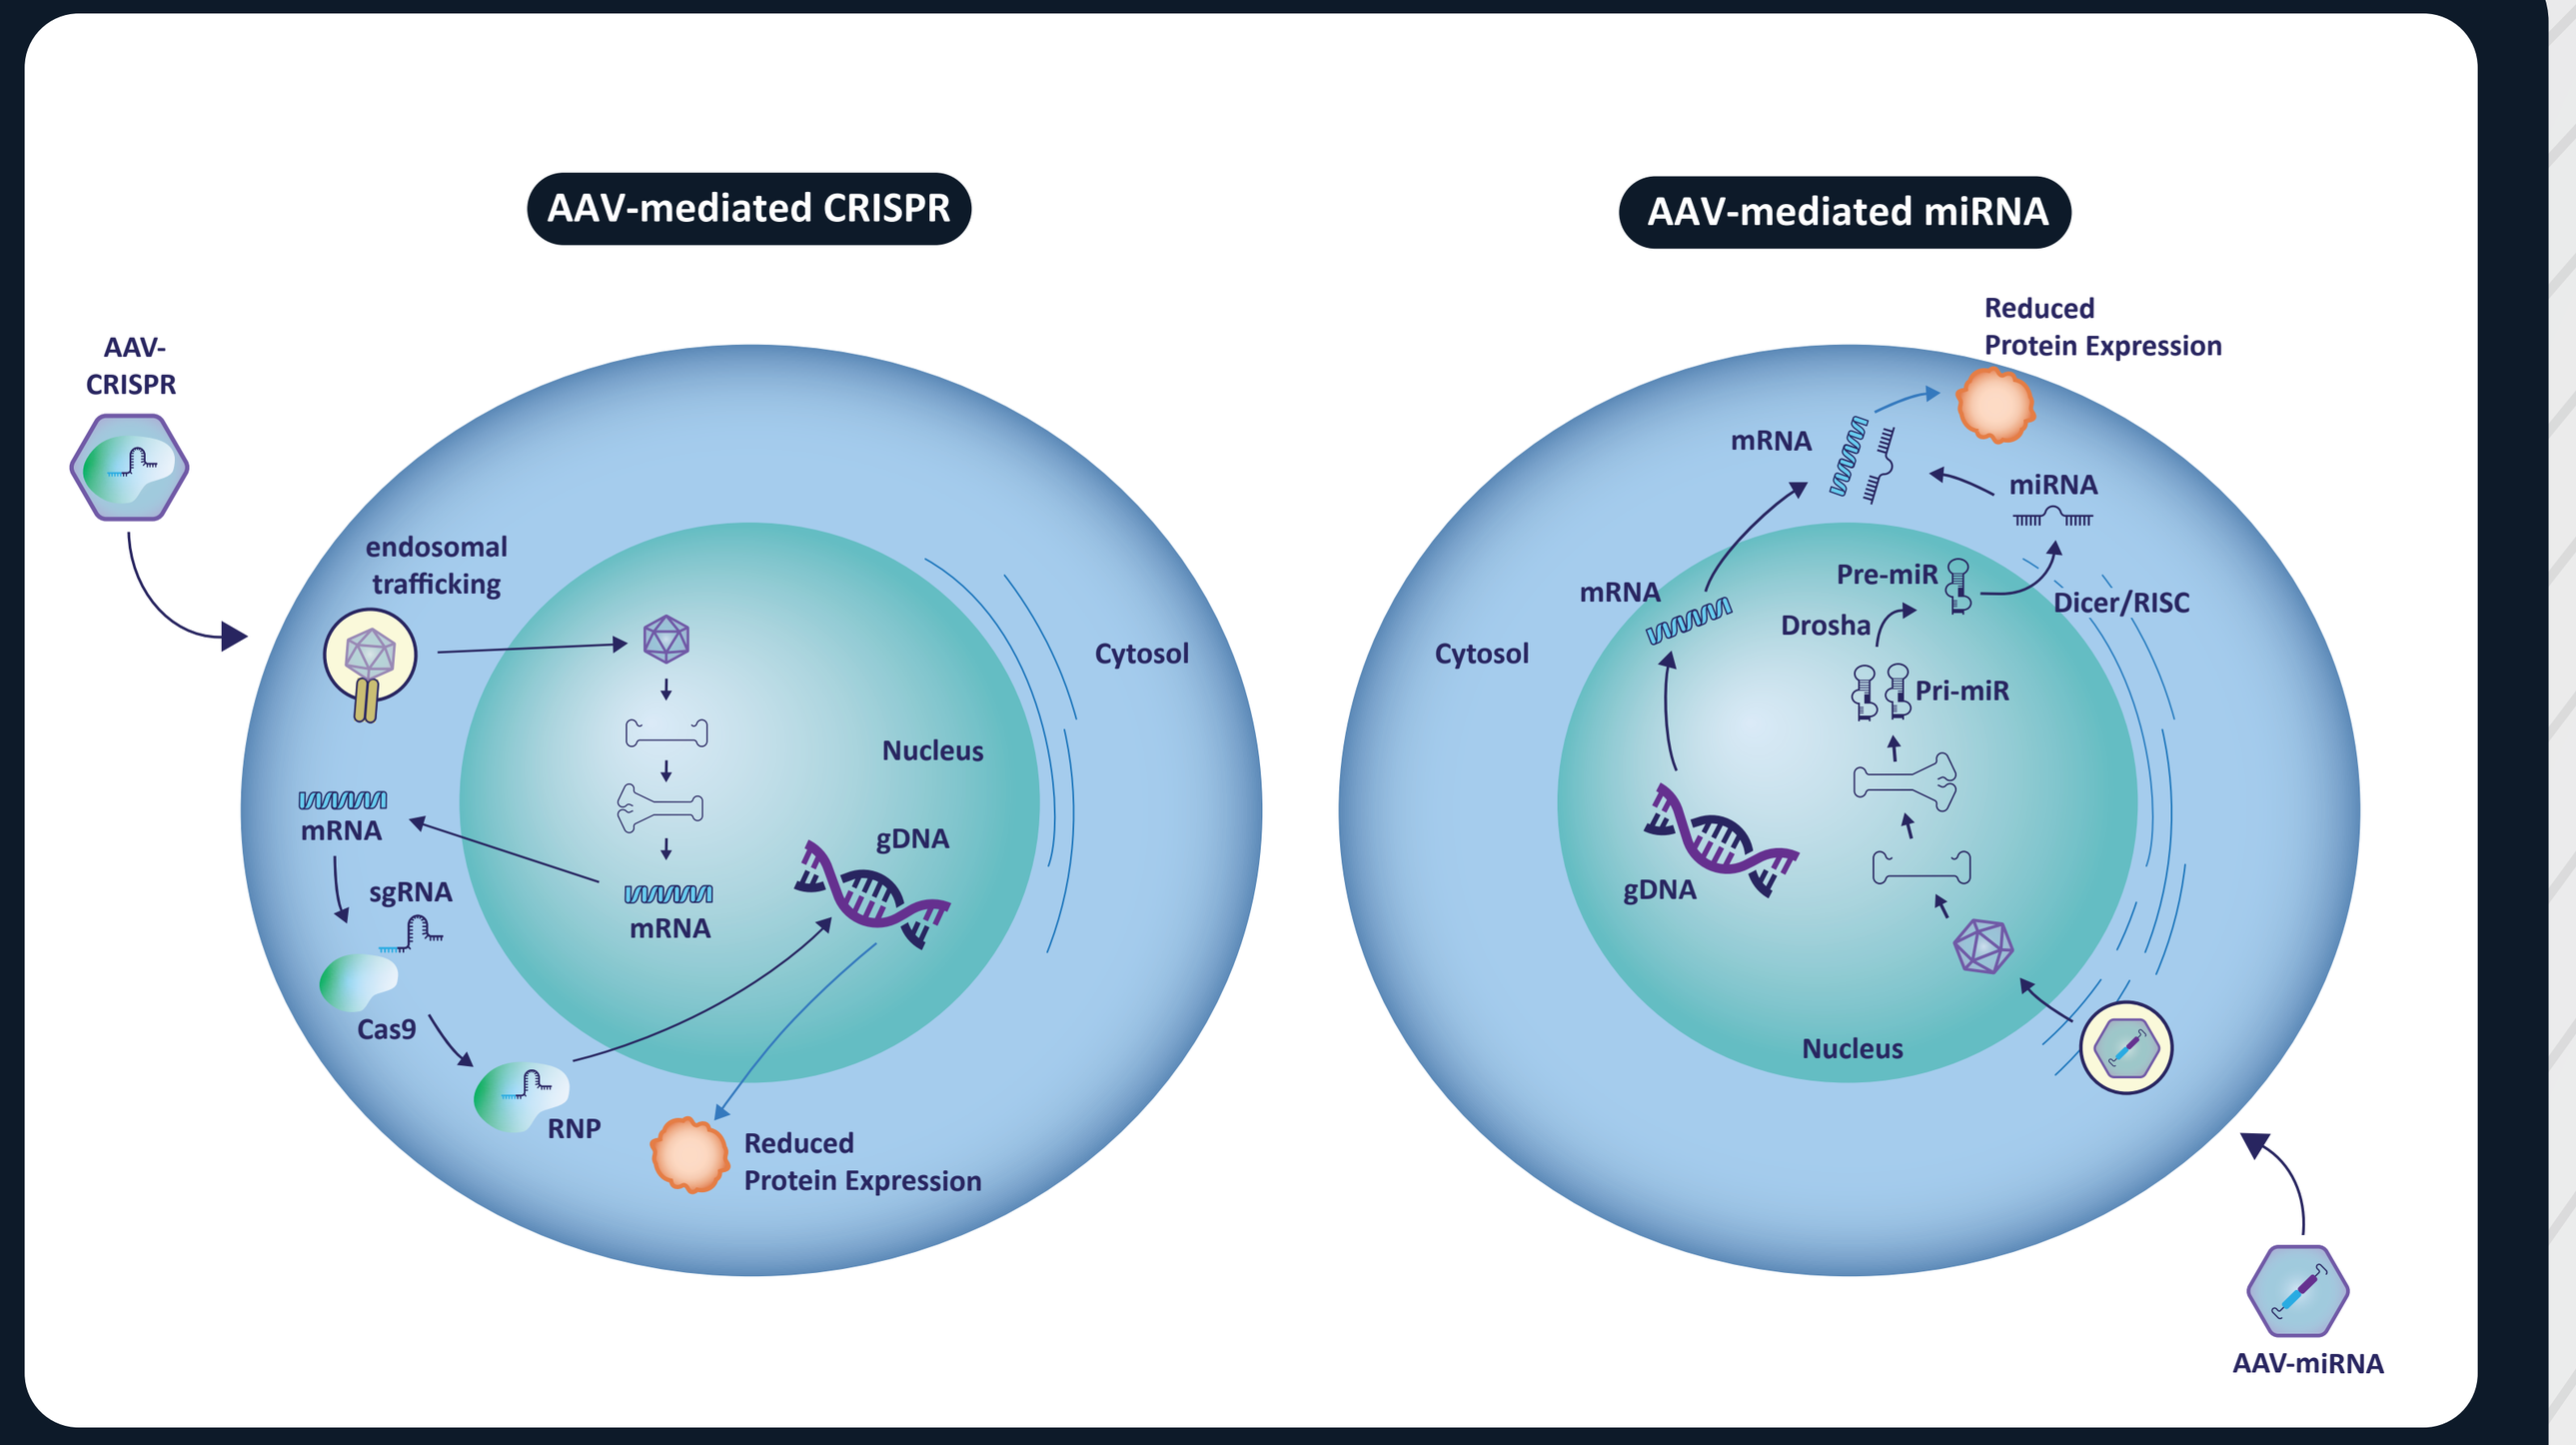
AAV-mediated CRISPR
AAV-mediated miRNA
AAV-CRISPR
endosomal trafficking
Cytosol
Nucleus
gDNA
mRNA
sgRNA
Cas9
RNP
Reduced Protein Expression
miRNA
Pre-miR
Drosha
Dicer/RISC
Pri-miR
AAV-miRNA

FIGURE 2: Dual Luciferase reporter readouts on miRNA screening

- Engineered microRNAs were designed using combinations of different microRNA scaffolds and Gene of Interest targeting sequences
- In total, more than 33 unique engineered microRNA plasmids were screened for their ability to reduce target gene levels as measured by a Dual Luciferase reporter assay
- The top 10 plasmids were chosen for further analysis (passenger strand generation, off-target knockdown)

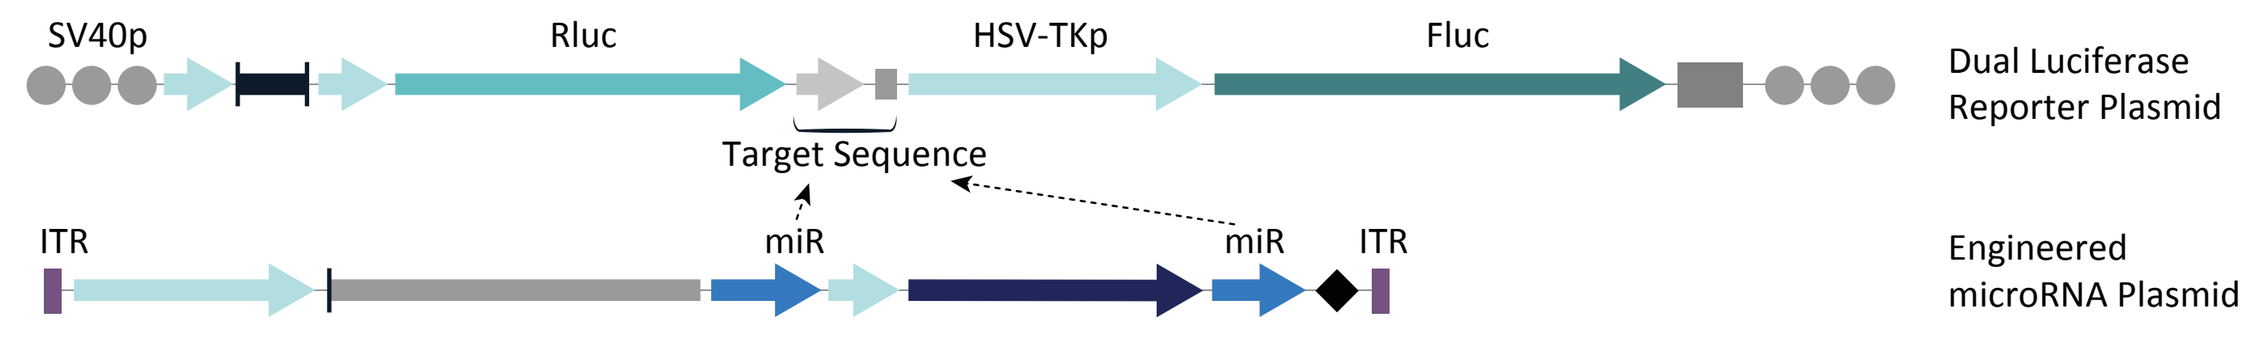
SV40p
Rluc
HSV-TKp
Fluc
Dual Luciferase Reporter Plasmid
Target Sequence
ITR
miR
miR
ITR
Engineered microRNA Plasmid

Legend: Luciferase assay-based screening of microRNA plasmids for guide strand activity (panels A-D), passenger strand activity (E), and off-target activity (F).

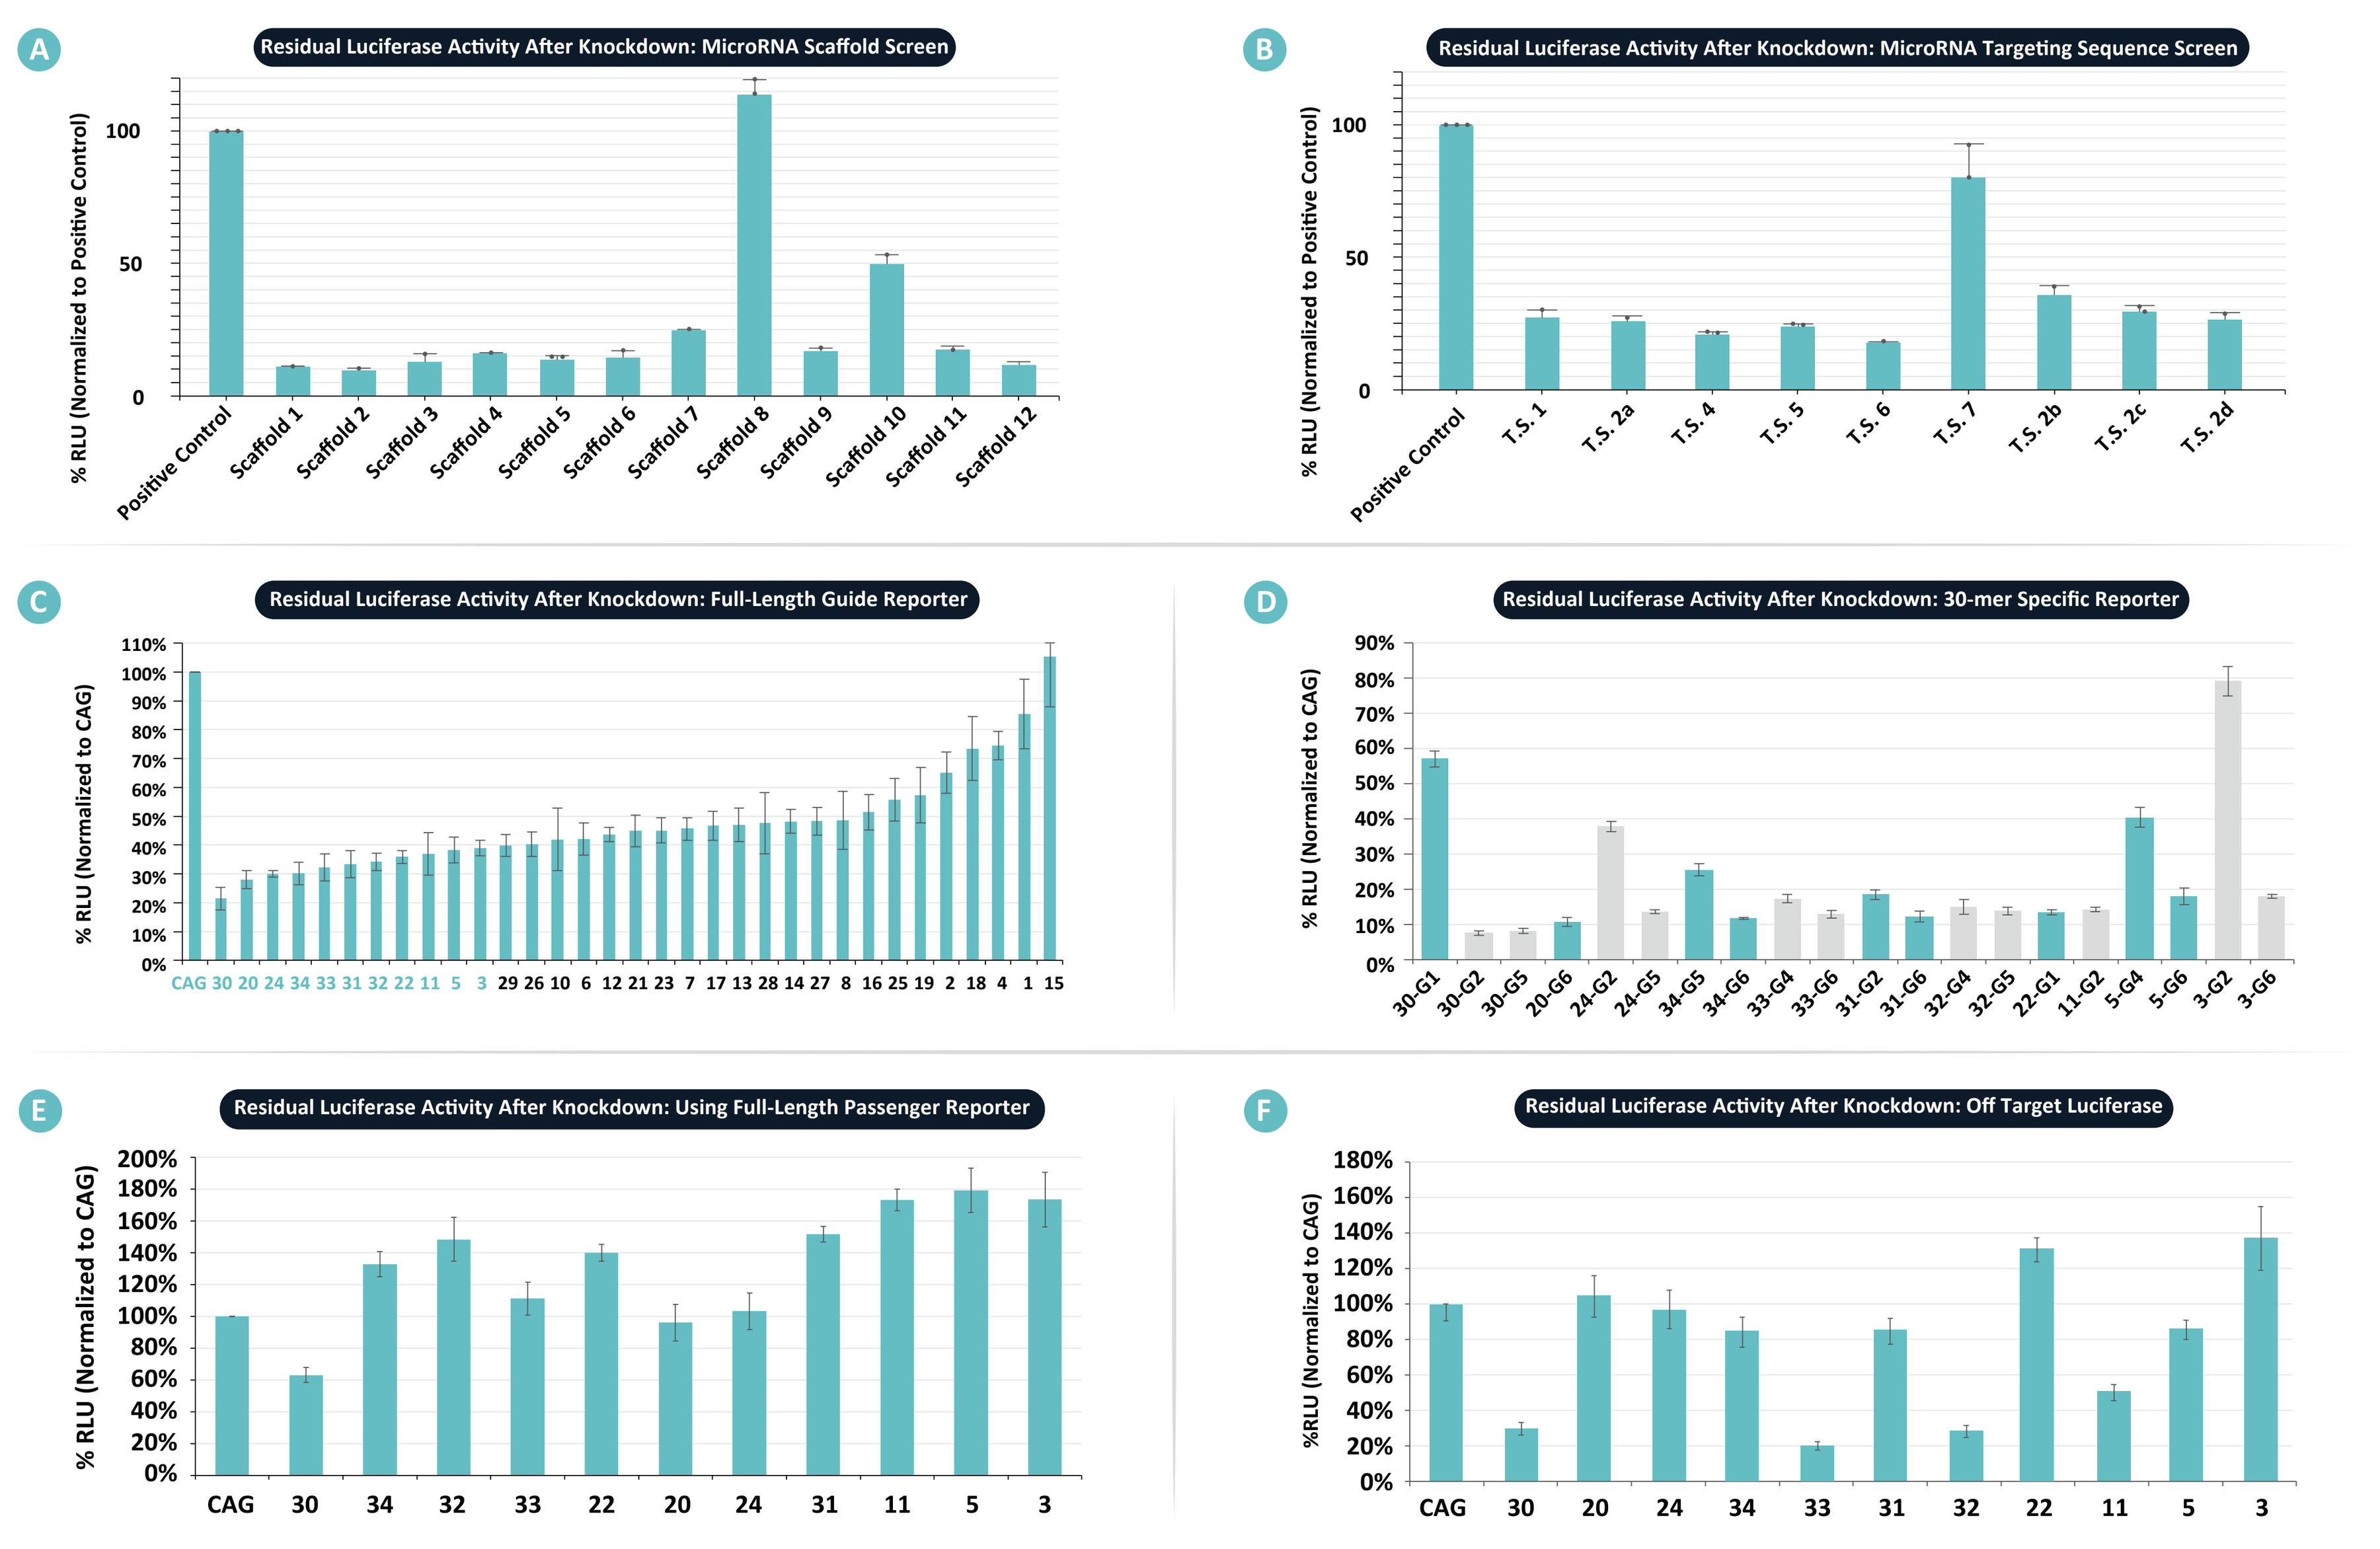
A
Residual Luciferase Activity After Knockdown: MicroRNA Scaffold Screen
% RLU (Normalized to Positive Control)
Positive Control
Scaffold 1
Scaffold 2
Scaffold 3
Scaffold 4
Scaffold 5
Scaffold 6
Scaffold 7
Scaffold 8
Scaffold 9
Scaffold 10
Scaffold 11
Scaffold 12
B
Residual Luciferase Activity After Knockdown: MicroRNA Targeting Sequence Screen
T.S. 1
T.S. 2a
T.S. 4
T.S. 5
T.S. 6
T.S. 7
T.S. 2b
T.S. 2c
T.S. 2d
C
Residual Luciferase Activity After Knockdown: Full-Length Guide Reporter
% RLU (Normalized to CAG)
CAG 30 20 24 34 33 31 32 22 11 5 3 29 26 10 6 12 21 23 7 17 13 28 14 27 8 16 25 19 2 18 4 1 15
D
Residual Luciferase Activity After Knockdown: 30-mer Specific Reporter
30-G1 30-G2 30-G5 20-G6 24-G2 24-G5 34-G5 34-G6 33-G4 33-G6 31-G2 31-G6 32-G4 32-G5 22-G1 11-G2 5-G4 5-G6 3-G2 3-G6
E
Residual Luciferase Activity After Knockdown: Using Full-Length Passenger Reporter
CAG 30 34 32 33 22 20 24 31 11 5 3
F
Residual Luciferase Activity After Knockdown: Off Target Luciferase
%RLU (Normalized to CAG)
CAG 30 20 24 34 33 31 32 22 11 5 3

FIGURE 3: NGS small RNA analysis of miRNA variants in vitro

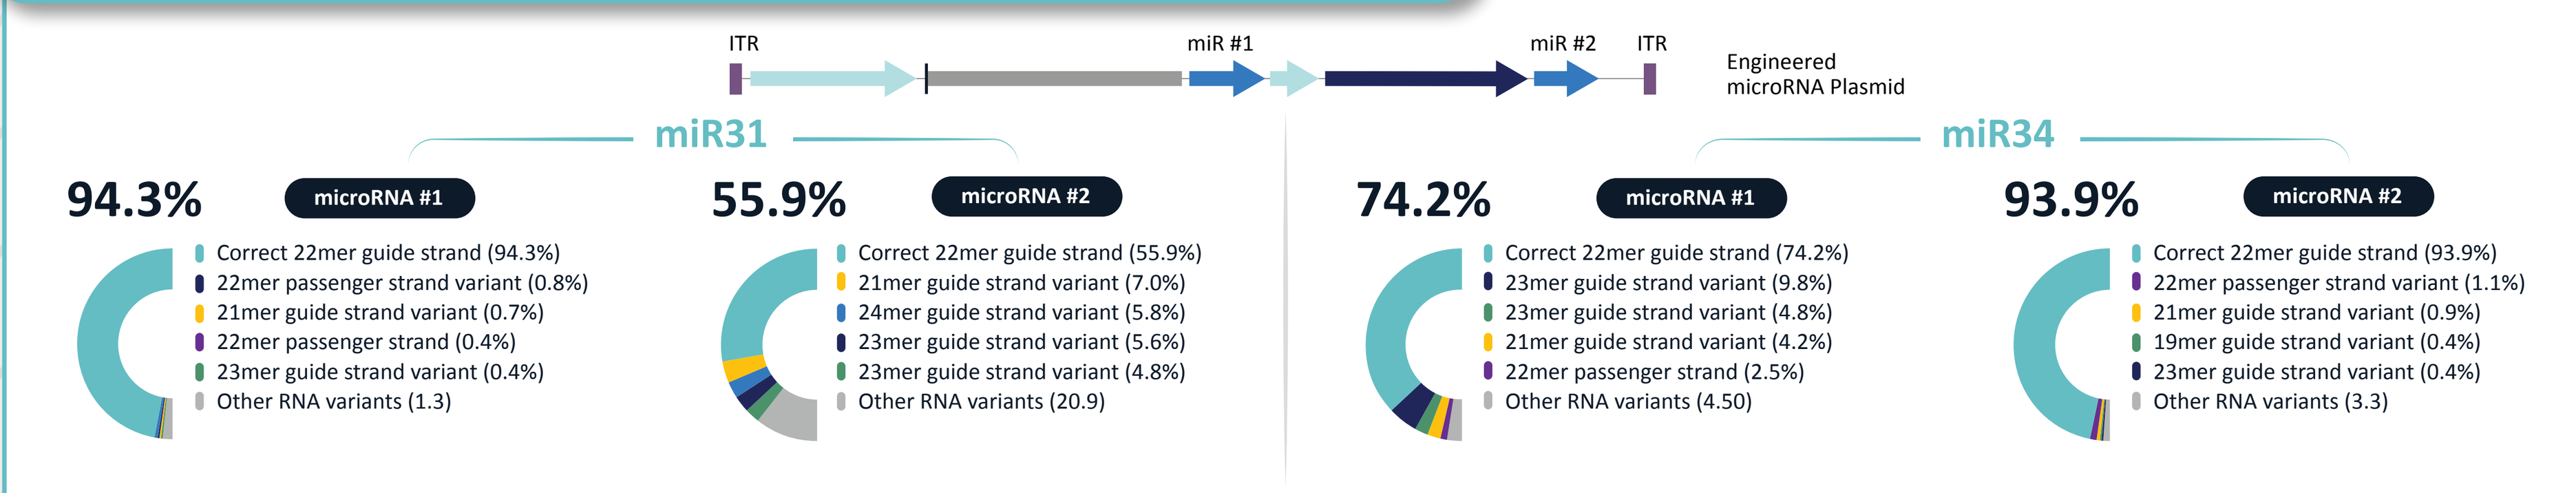
ITR
miR #1
miR #2
ITR
Engineered microRNA Plasmid
miR31
94.3%
microRNA #1
Correct 22mer guide strand (94.3%)
22mer passenger strand variant (0.8%)
21mer guide strand variant (0.7%)
22mer passenger strand (0.4%)
23mer guide strand variant (0.4%)
Other RNA variants (1.3)
55.9%
microRNA #2
Correct 22mer guide strand (55.9%)
21mer guide strand variant (7.0%)
24mer guide strand variant (5.8%)
23mer guide strand variant (5.6%)
23mer guide strand variant (4.8%)
Other RNA variants (20.9)
miR34
74.2%
microRNA #1
Correct 22mer guide strand (74.2%)
23mer guide strand variant (9.8%)
23mer guide strand variant (4.8%)
21mer guide strand variant (4.2%)
22mer passenger strand (2.5%)
Other RNA variants (4.50)
93.9%
microRNA #2
Correct 22mer guide strand (93.9%)
22mer passenger strand variant (1.1%)
21mer guide strand variant (0.9%)
19mer guide strand variant (0.4%)
23mer guide strand variant (0.4%)
Other RNA variants (3.3)

Legend: Small RNA-seq of microRNA plasmids to evaluate efficacy of guide and passenger strand generation

- MicroRNAs within the miR31 and miR34 plasmids are processed to produce predominantly the correct 22mer guide strand sequence

FIGURE 4: In vitro analysis of CRISPR and miRNA activity

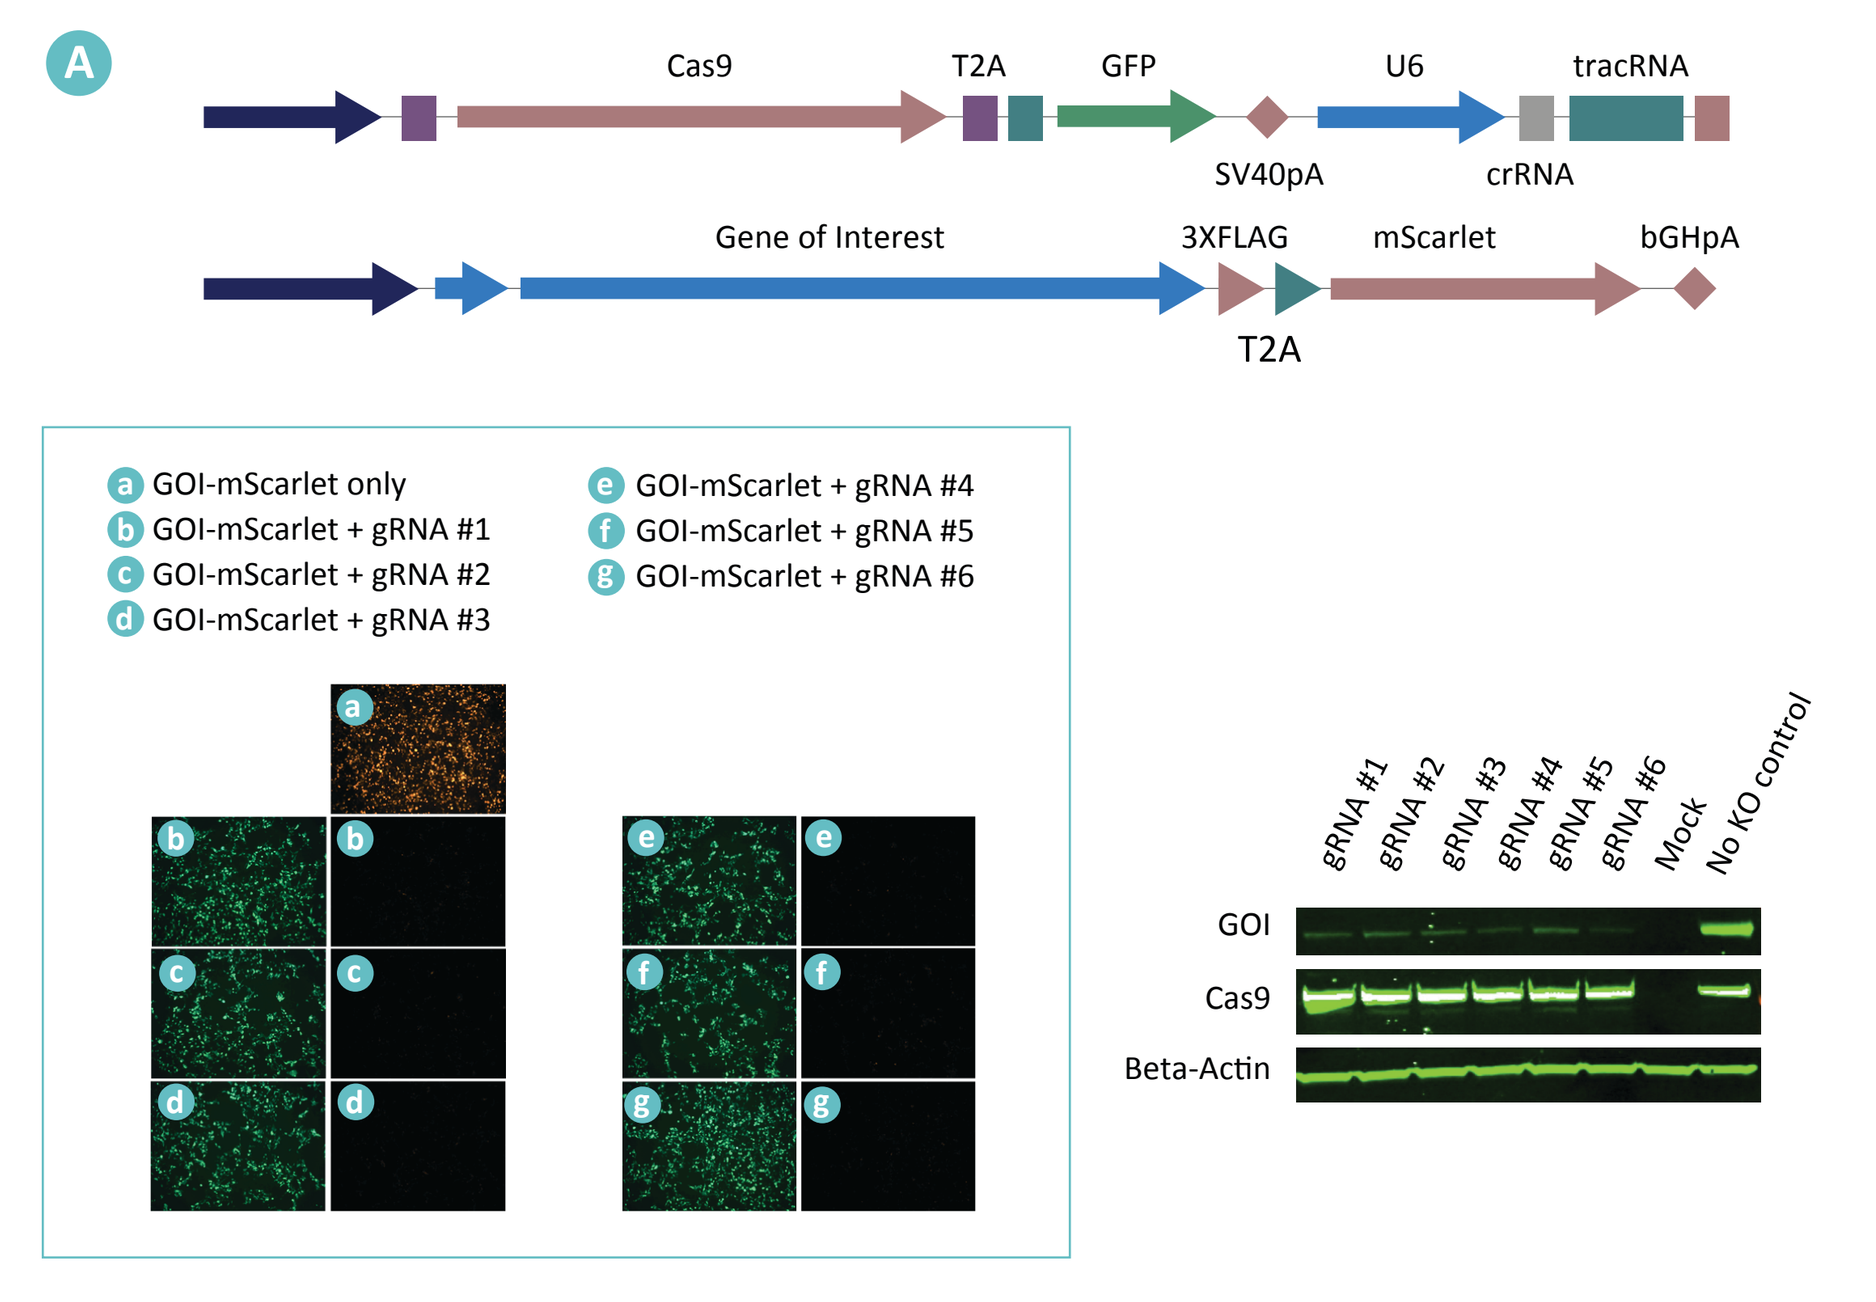
A
Cas9
T2A
GFP
U6
tracRNA
SV40pA
crRNA
Gene of Interest
3XFLAG
mScarlet
bGHpA
T2A
a GOI-mScarlet only
b GOI-mScarlet + gRNA #1
c GOI-mScarlet + gRNA #2
d GOI-mScarlet + gRNA #3
e GOI-mScarlet + gRNA #4
f GOI-mScarlet + gRNA #5
g GOI-mScarlet + gRNA #6
gRNA #1
gRNA #2
gRNA #3
gRNA #4
gRNA #5
gRNA #6
Mock
No KO control
GOI
Cas9
Beta-Actin

- Six different guide RNAs were tested for efficacy in reducing protein levels of the target Gene of Interest via fluorescence imaging and Western Blot
- GFP fragment is removed for vector production

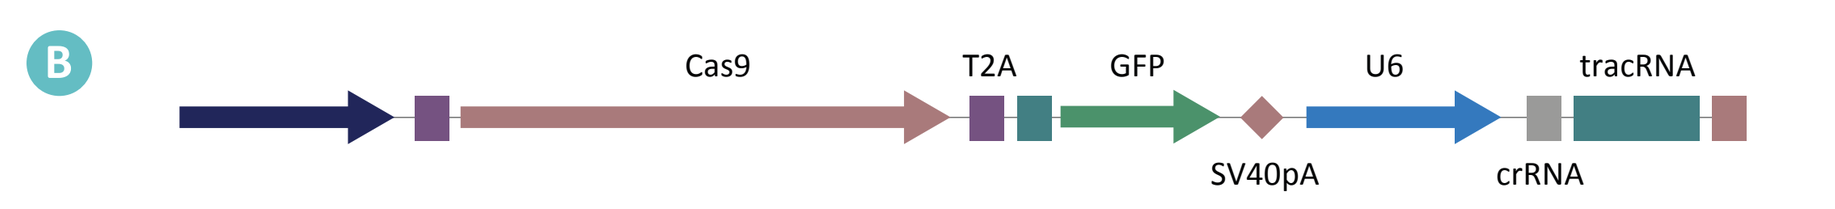
B
Cas9
T2A
GFP
U6
tracRNA
SV40pA
crRNA

Indel Position	-5	-4	-3	-2	-1	0	+1	Overall Editing Efficiency
gRNA #1	<1%	3.1%	3.5%	<1%	5.9%	46.7%	9.3%	31.1%
gRNA #2	<1%	6.3%	<1%	7.6%	4.3%	40.9%	8.9%	50.6%
gRNA #3	<1%	<1%	4.9%	2.3%	9.7%	60.4%	10.1%	36.1%
gRNA #4	<1%	<1%	5%	2.3%	9.6%	60.4%	10%	36.0%
gRNA #5	<1%	<1%	<1%	8.2%	2.1%	57.7%	7.9%	39.7%
gRNA #6	<1%	5.1%	<1%	9.8%	6.9%	49.0%	<1%	42.1%

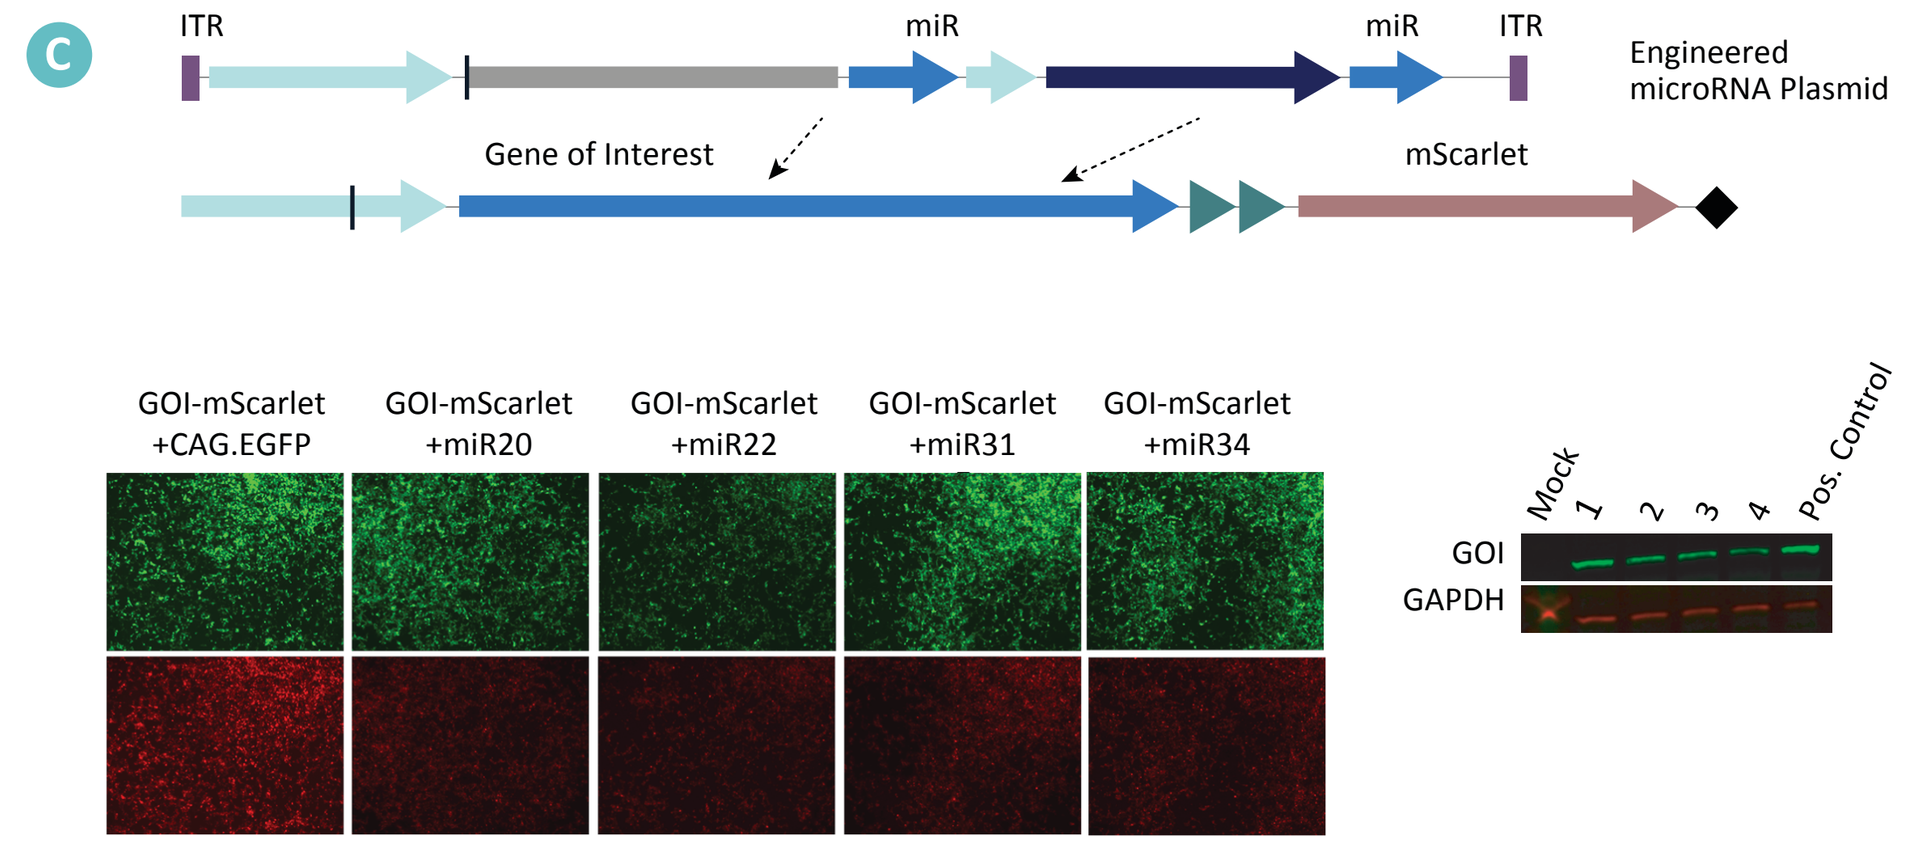
C
ITR
miR
miR
ITR
Engineered microRNA Plasmid
Gene of Interest
mScarlet
GOI-mScarlet +CAG.EGFP
GOI-mScarlet +miR20
GOI-mScarlet +miR22
GOI-mScarlet +miR31
GOI-mScarlet +miR34
Mock
1
2
3
4
Pos. Control
GOI
GAPDH

Legend: (A) Schematics of CRISPR/Cas9 plasmids used for in vitro screening to observe protein reduction via fluorescence imaging and Western Blot; (B) Efficiency of DNA editing and indel creation using TIDE analysis; (C) Schematics of microRNA plasmids used for in vitro screening to observe protein reduction via fluorescence imaging and Western Blot.

FIGURE 5: Codon-modification of add-back gene resists reduction of expression by miRNA and CRISPR plasmids

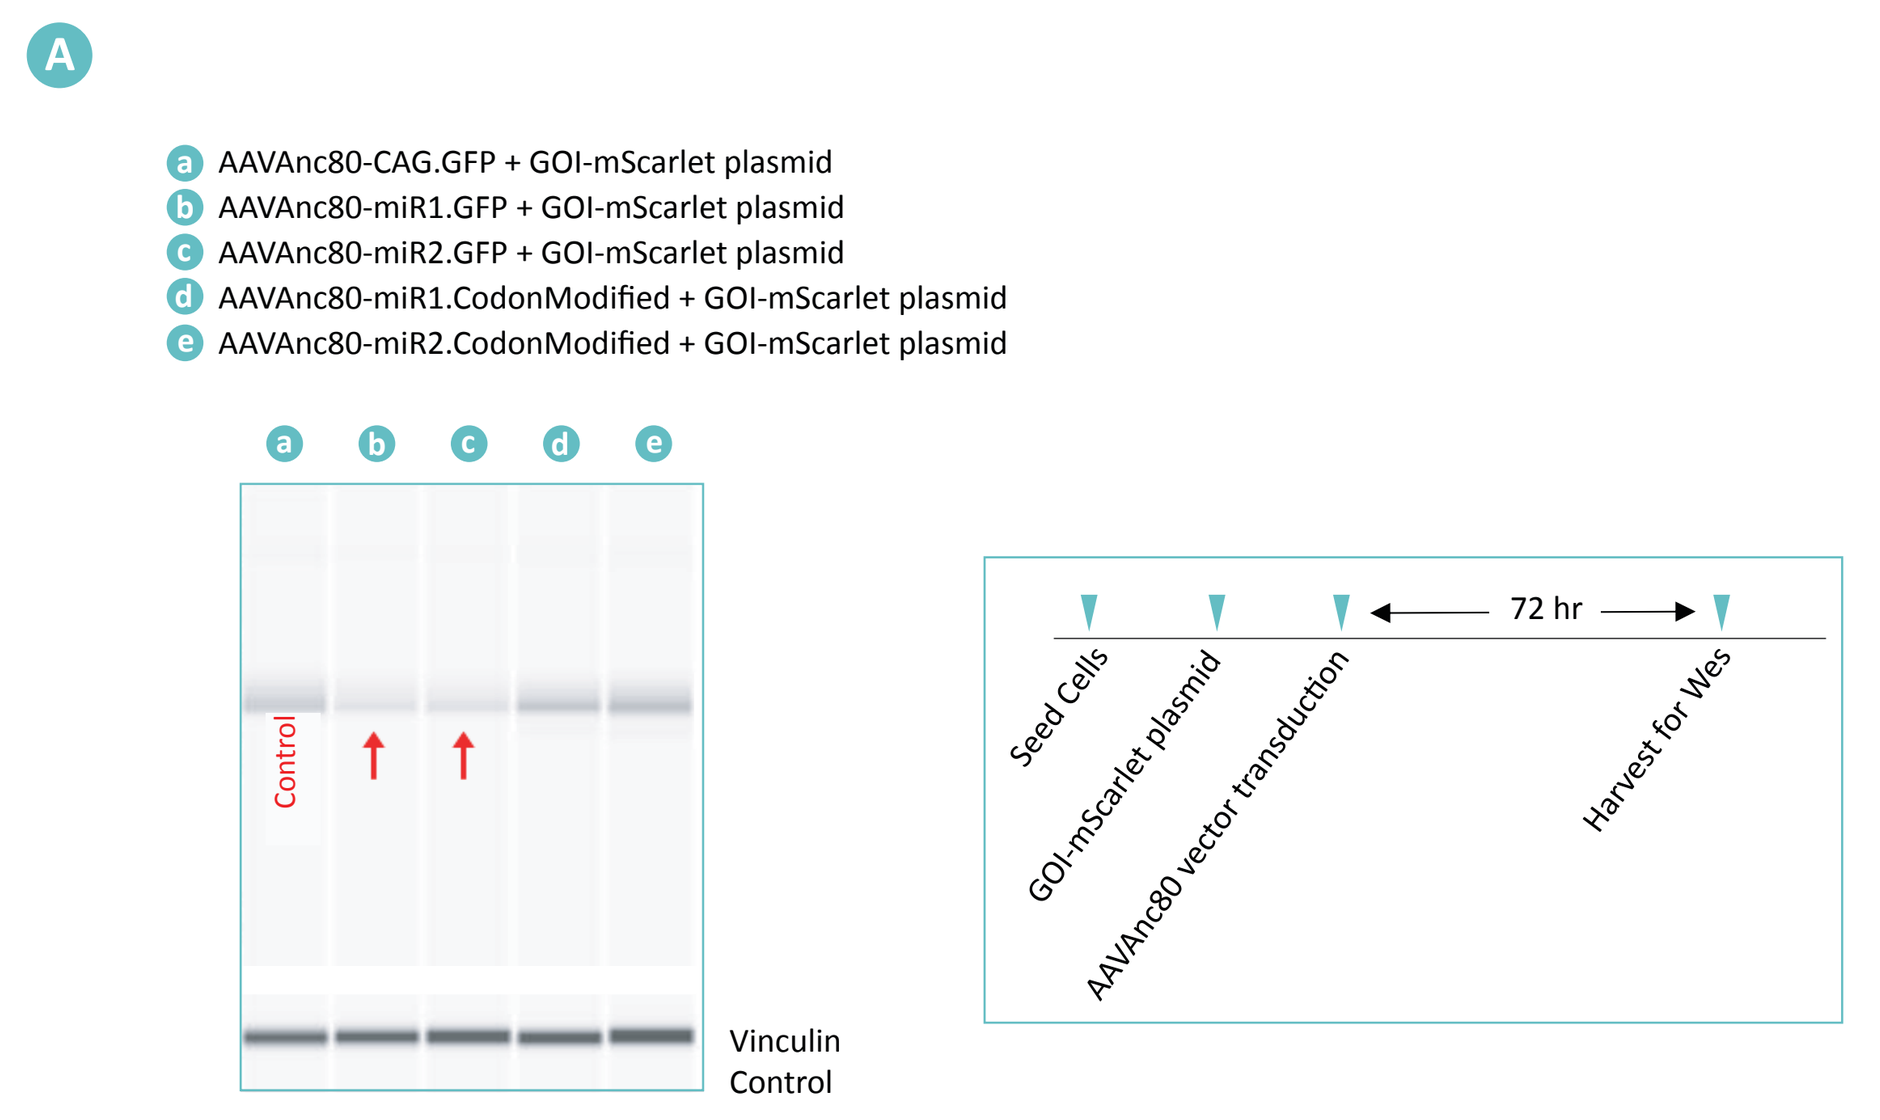
A
a AAVAnc80-CAG.GFP + GOI-mScarlet plasmid
b AAVAnc80-miR1.GFP + GOI-mScarlet plasmid
c AAVAnc80-miR2.GFP + GOI-mScarlet plasmid
d AAVAnc80-miR1.CodonModified + GOI-mScarlet plasmid
e AAVAnc80-miR2.CodonModified + GOI-mScarlet plasmid
Control
Vinculin Control
Seed Cells
GOI-mScarlet plasmid
AAVAnc80 vector transduction
72 hr
Harvest for Wes

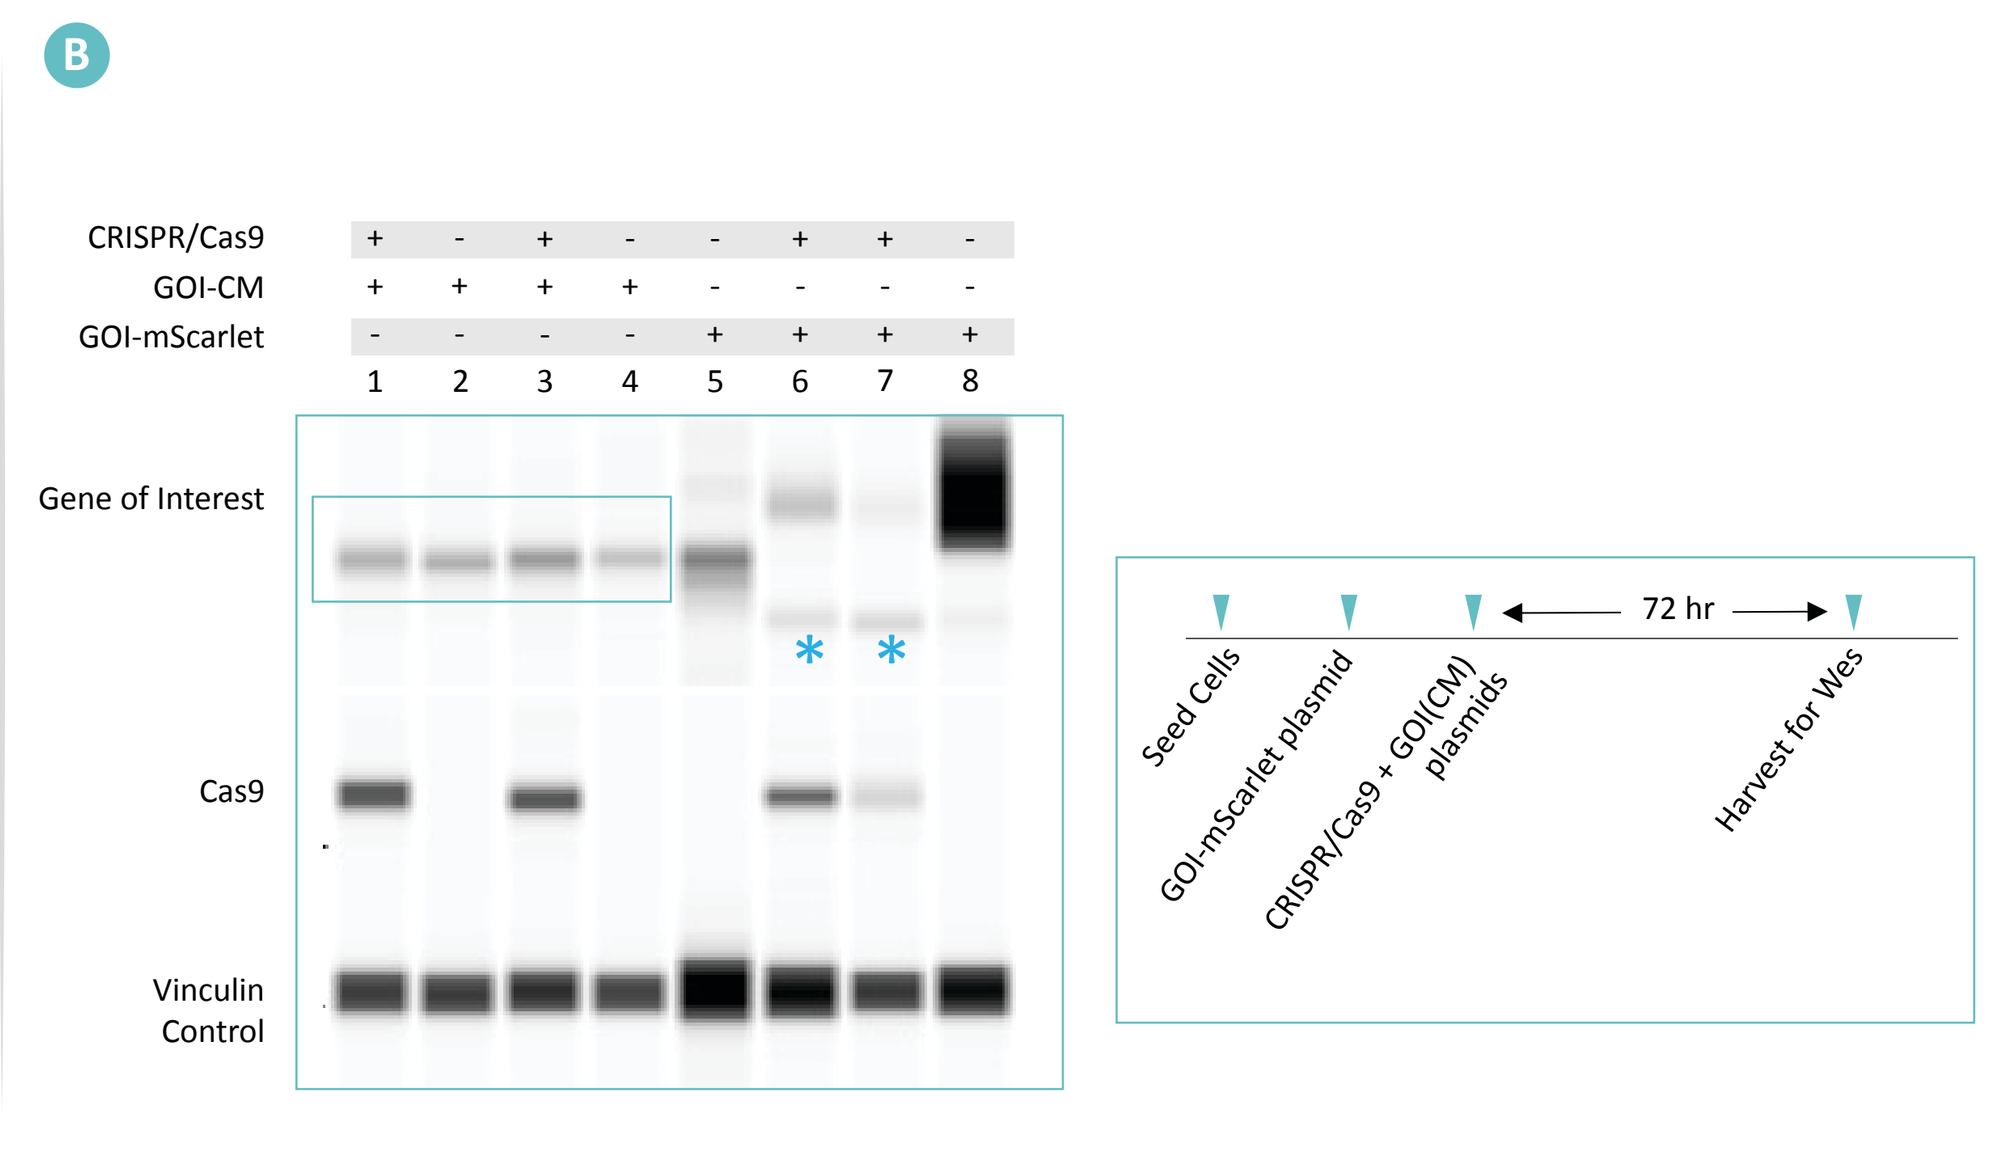
B
CRISPR/Cas9
GOI-CM
GOI-mScarlet
1 2 3 4 5 6 7 8
Gene of Interest
Cas9
Vinculin Control
Seed Cells
GOI-mScarlet plasmid
CRISPR/Cas9 + GOI(CM) plasmids
72 hr
Harvest for Wes

Legend: Western gel images showing that codon-modified versions of the Gene of Interest are resistant to (A) microRNA knockdown effects using two different microRNA vectors and (B) CRISPR/Cas9-mediated knockout effects using two different guide RNA plamids

CONCLUSION

- Our results, both with RNAi gene silencing and with CRISPR/Cas9 gene editing, build foundational platform capabilities which may be used to develop potential future AAV vector-mediated therapies for many types of ADHL and other inner ear conditions

AKOUOS